First in human phase 1/2a study of PEN-221 somatostatin analog-DM1 conjugate for patients with advanced neuroendocrine tumors or small cell lung cancer: Phase 1 results

Abstract #4097

ML Johnson, T Meyer, DM Halperin, AT Fojo, N Cook, LS Blaszkowsky, BL Schlechter, JC Yao, Y Jemiai, K Kriksciukaite, M Jones, N Ezema, L Mei, L Alland, MH Kulke

Sarah Cannon Research Institute, Nashville, TN; University College London Cancer Institute, London, United Kingdom; University of Texas MD Anderson Cancer Center, Houston, TX; Columbia University Medical Center, New York, NY; University of Manchester, Manchester, United Kingdom; Massachusetts General Hospital, Boston, MA; Beth Israel Deaconess Medical Center, Boston, MA; Cytel, Cambridge, MA; Tarveda Therapeutics, Watertown, MA; Syneos Health, Raleigh, NC; Tarveda Therapeutics, Watertown, US; Boston University School of Medicine, Boston, MA

PEN-221

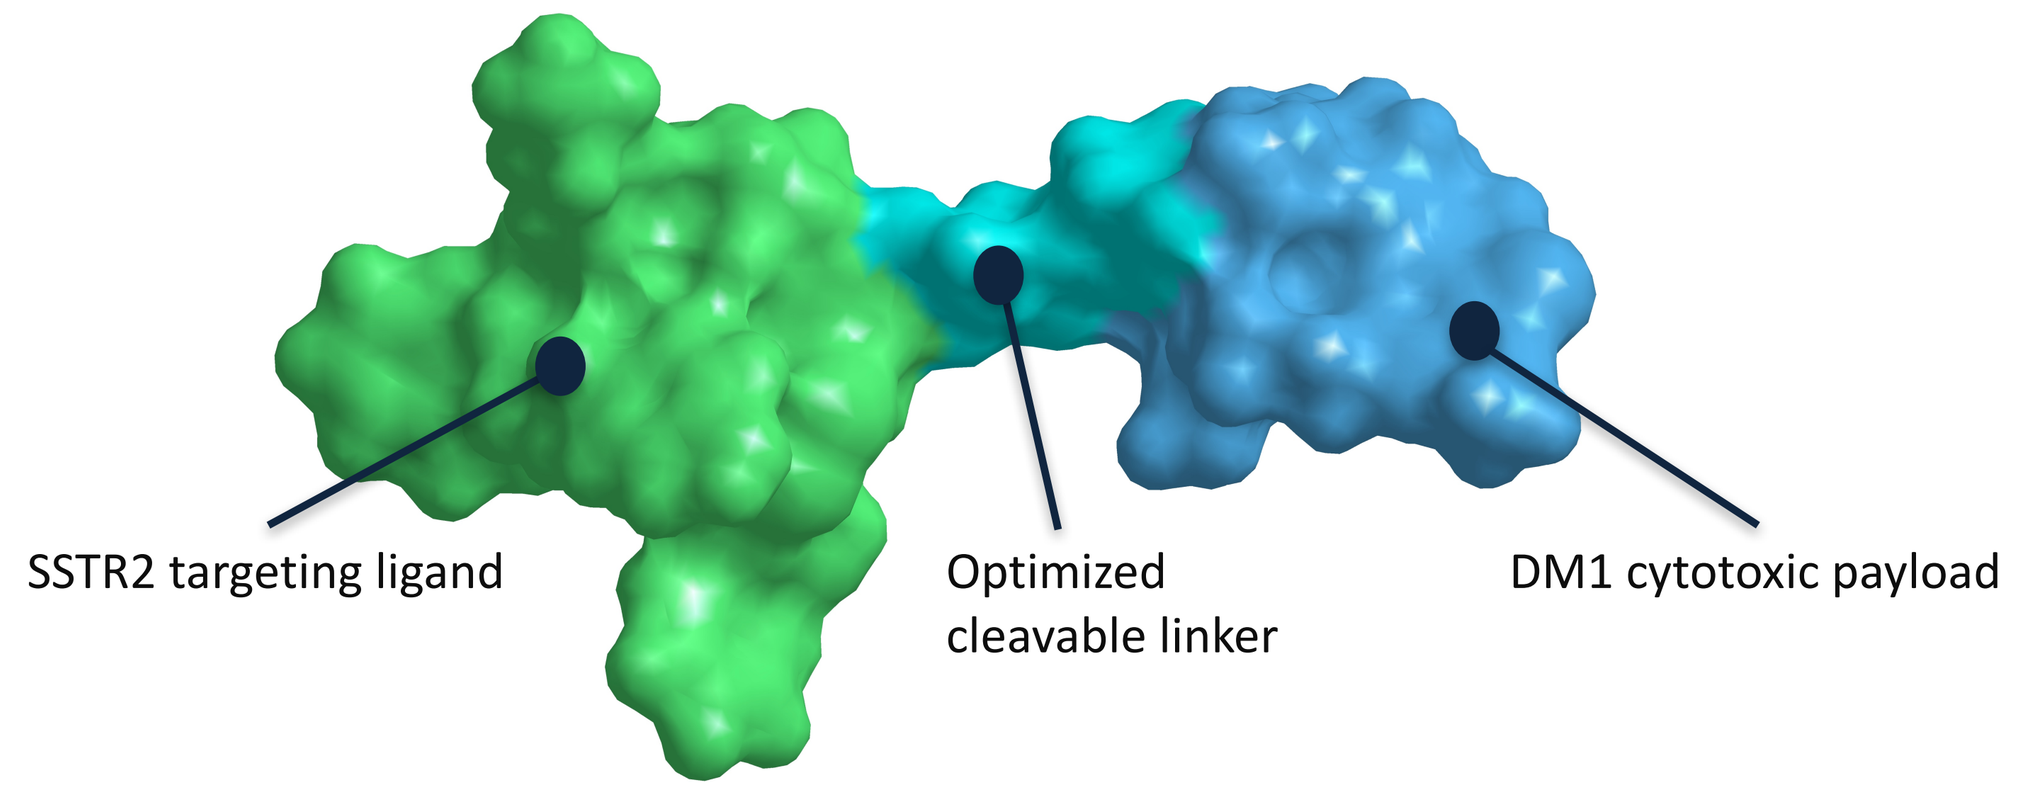
SSTR2 targeting ligand
Optimized cleavable linker
DM1 cytotoxic payload

PEN-221 is a miniature drug conjugate consisting of a peptide ligand that is highly selective in targeting the somatostatin receptor 2 (SSTR2), joined through a cleavable linker to the potent cytotoxic payload DM1. SSTR2 is over expressed on the cell surface of solid tumors including Neuroendocrine Tumors (NETs), Small Cell Lung Cancers (SCLC) and metastatic castrate resistant prostate cancer. There remains an unmet medical need in these patient populations. In non-clinical experiments, PEN-221 binds with high affinity and selectivity to SSTR2. On binding, PEN-221 triggers SSTR2 internalization resulting in the accumulation of the DM1 payload in tumor cells followed by cell cycle arrest and apoptosis. PEN-221 achieves complete and durable regressions in multiple SSTR2 xenograft mouse models.

Study Objectives

- The primary objective of this Phase 1 study was to investigate the safety and tolerability, determine the maximum tolerated dose (MTD), and Recommended Phase 2 Dose (RP2D) of PEN-221 when administered IV on an every 3 week schedule in patients with SSTR2-expressing advanced cancers, including advanced gastroenteropancreatic (GEP) or lung or thymus or other NETs or SCLC or LCNEC of the lung (ClinicalTrials.gov Identifier: NCT02936323)
- Secondary objectives included characterizing the safety and tolerability of PEN-221, including both acute and chronic toxicities, measuring the plasma pharmacokinetics (PK) of PEN-221, DM1, and peptide from PEN-221, and assessing preliminary anti-tumor activity of PEN-221 as defined by duration of response and RECIST 1.1
- Exploratory objectives included assessing biomarker changes that include chromogranin A (CgA), neuron-specific enolase (NSE), and circulating tumor cells (CTCs) in the blood

Study design

- All patients provided written informed consent for pre-screening and subsequently for participation in the study
- Tumor SSTR2 expression was determined by imaging within the preceding approximately 6 months, with Octreoscan or ^{68}Ga-DOTATATE, ^{68}Ga-DOTATOC, or ^{68}Ga-DOTANOC.
- PEN-221 was administered as a 1 hour IV infusion once on an every 3 week schedule to escalating cohorts of 2-6 patients
- Safety was assessed during the study by vital sign measurements, physical examinations, neurological examinations, ECOG PS, documentation of adverse events (AEs), clinical laboratory tests, and ECGs
- Serial PK samples were collected prior, during and up to 10 hours after the start of infusion on Day 1 of Cycle 1 and Cycle 3. Samples were analyzed for PEN-221, Unconjugated DM1, Unconjugated SSTR2 targeting peptide and free sulfhydryl DM1.
- Disease response was assessed by duration of response and RECIST 1.1
- An adaptive Bayesian Logistic Regression Model (BLRM) guided by the Escalation with Overdose Control (EWOC) principle (Babb et al; Neuenschwander et al) was employed to make dose recommendations and estimate the MTD, defined as the dose with the highest posterior probability of targeted toxicity (16%-33%) that fulfills the EWOC (probability > 25% of excessive toxicity, i.e. greater than 33%). Intra-patient dose escalation was permitted

SSTR2 Imaging

- All patients were required to have SSTR2 expressing tumors

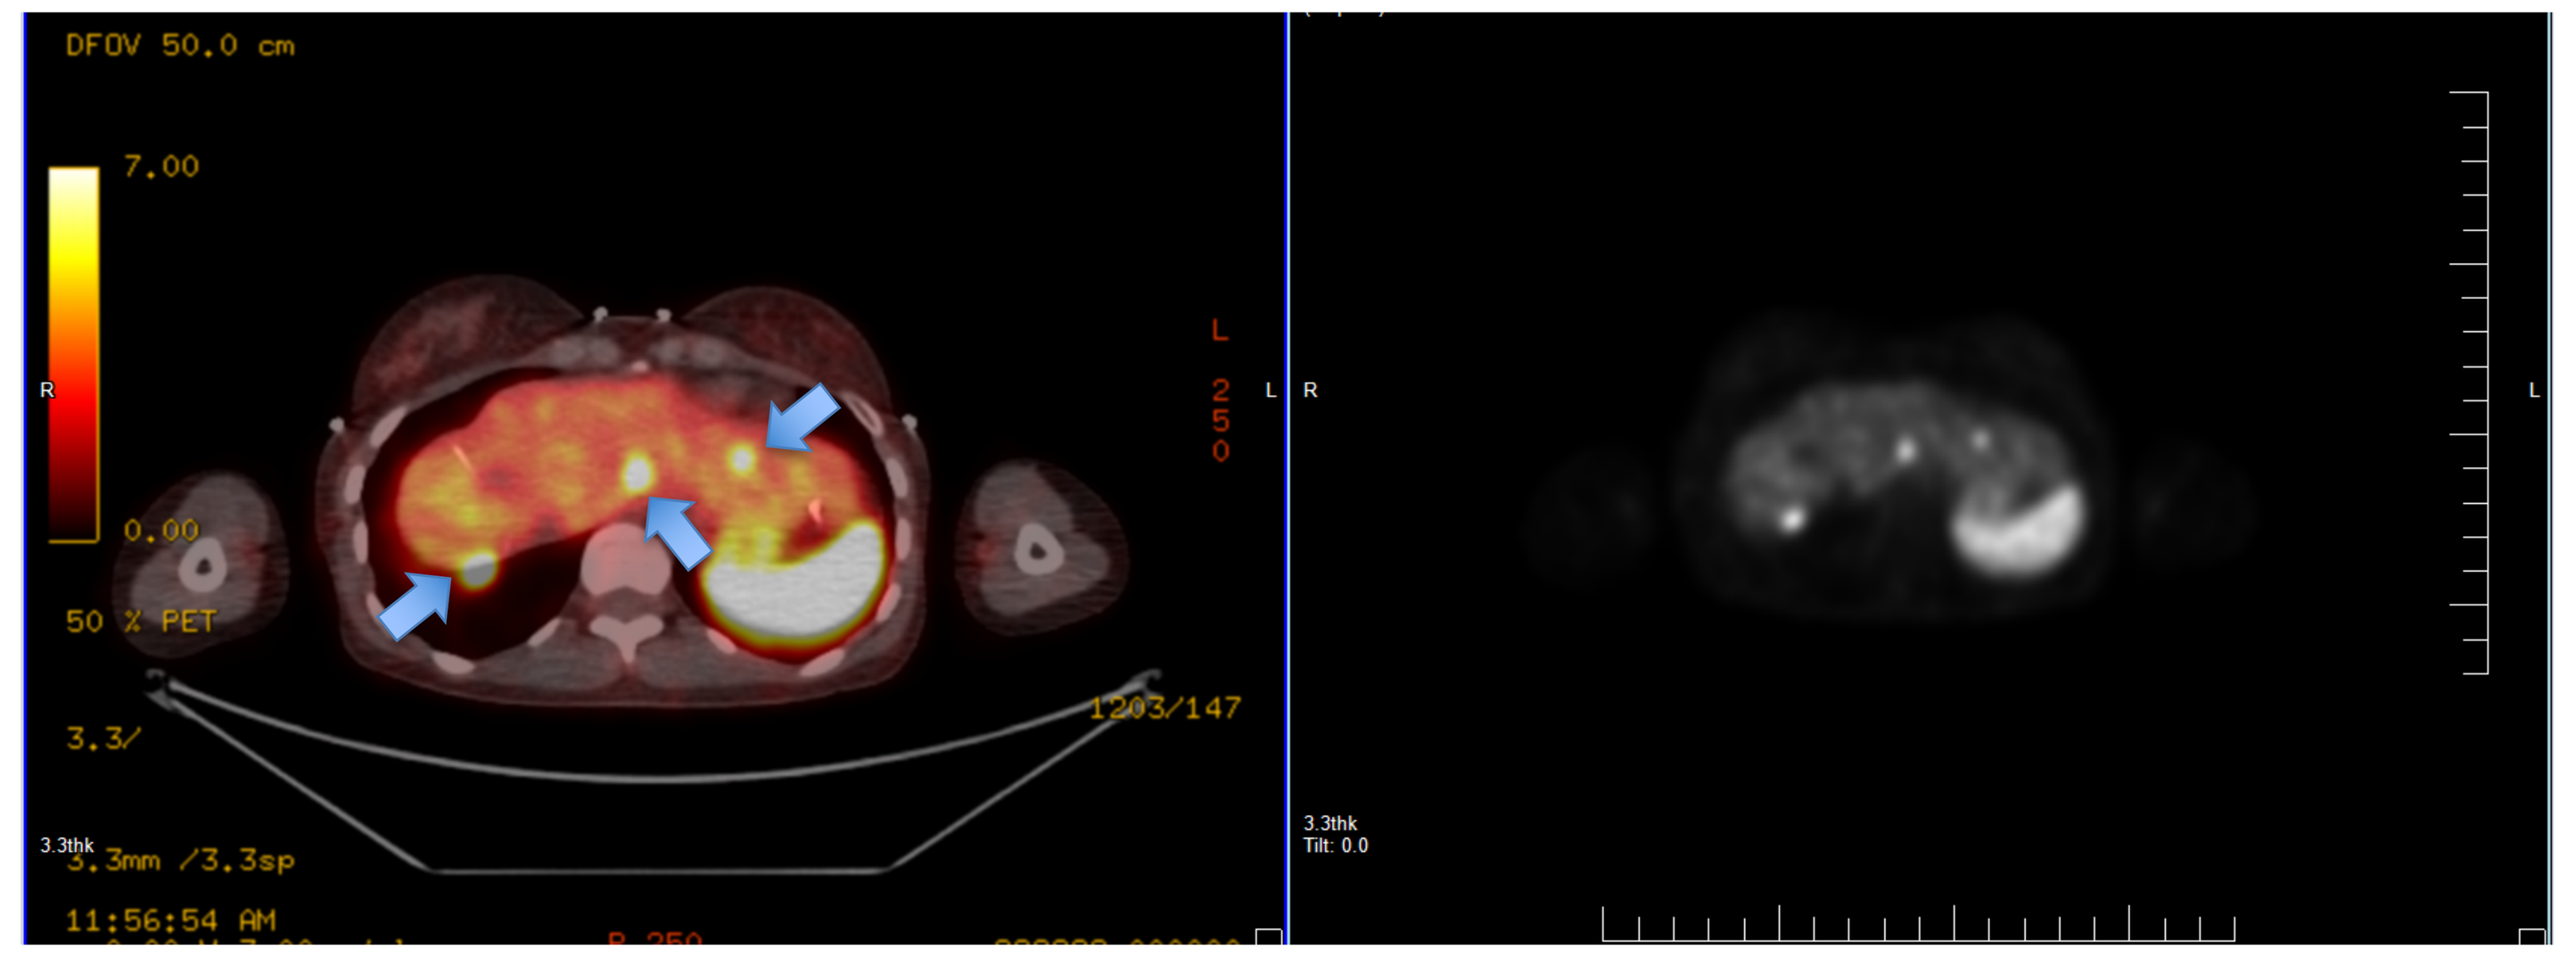
DFOV 50.0 cm
7.00
0.00
50 % PET
11:56:54 AM

Image from a ^{68}Ga-DOTATATE scan of a 30 year old woman with metastatic well-differentiated low grade mid-ileal NET, showing abnormal radiopharmaceutical uptake of in 3 liver nodules (arrowed) consistent with SSTR-containing metastases. High uptake in the spleen (right) is physiologic.

Patient Characteristics And Dose Escalation

Characteristic	Data
Sex	13M, 10F
Age, median (range)	61, (27 – 74)
Tumor types	
GI NET	9
PNET	5
Lung NET	5
Pheochromocytoma	2
NET of Unknown Primary	1
Small Cell Lung Cancer	1
Prior therapies, median (range)	3, (1-8)

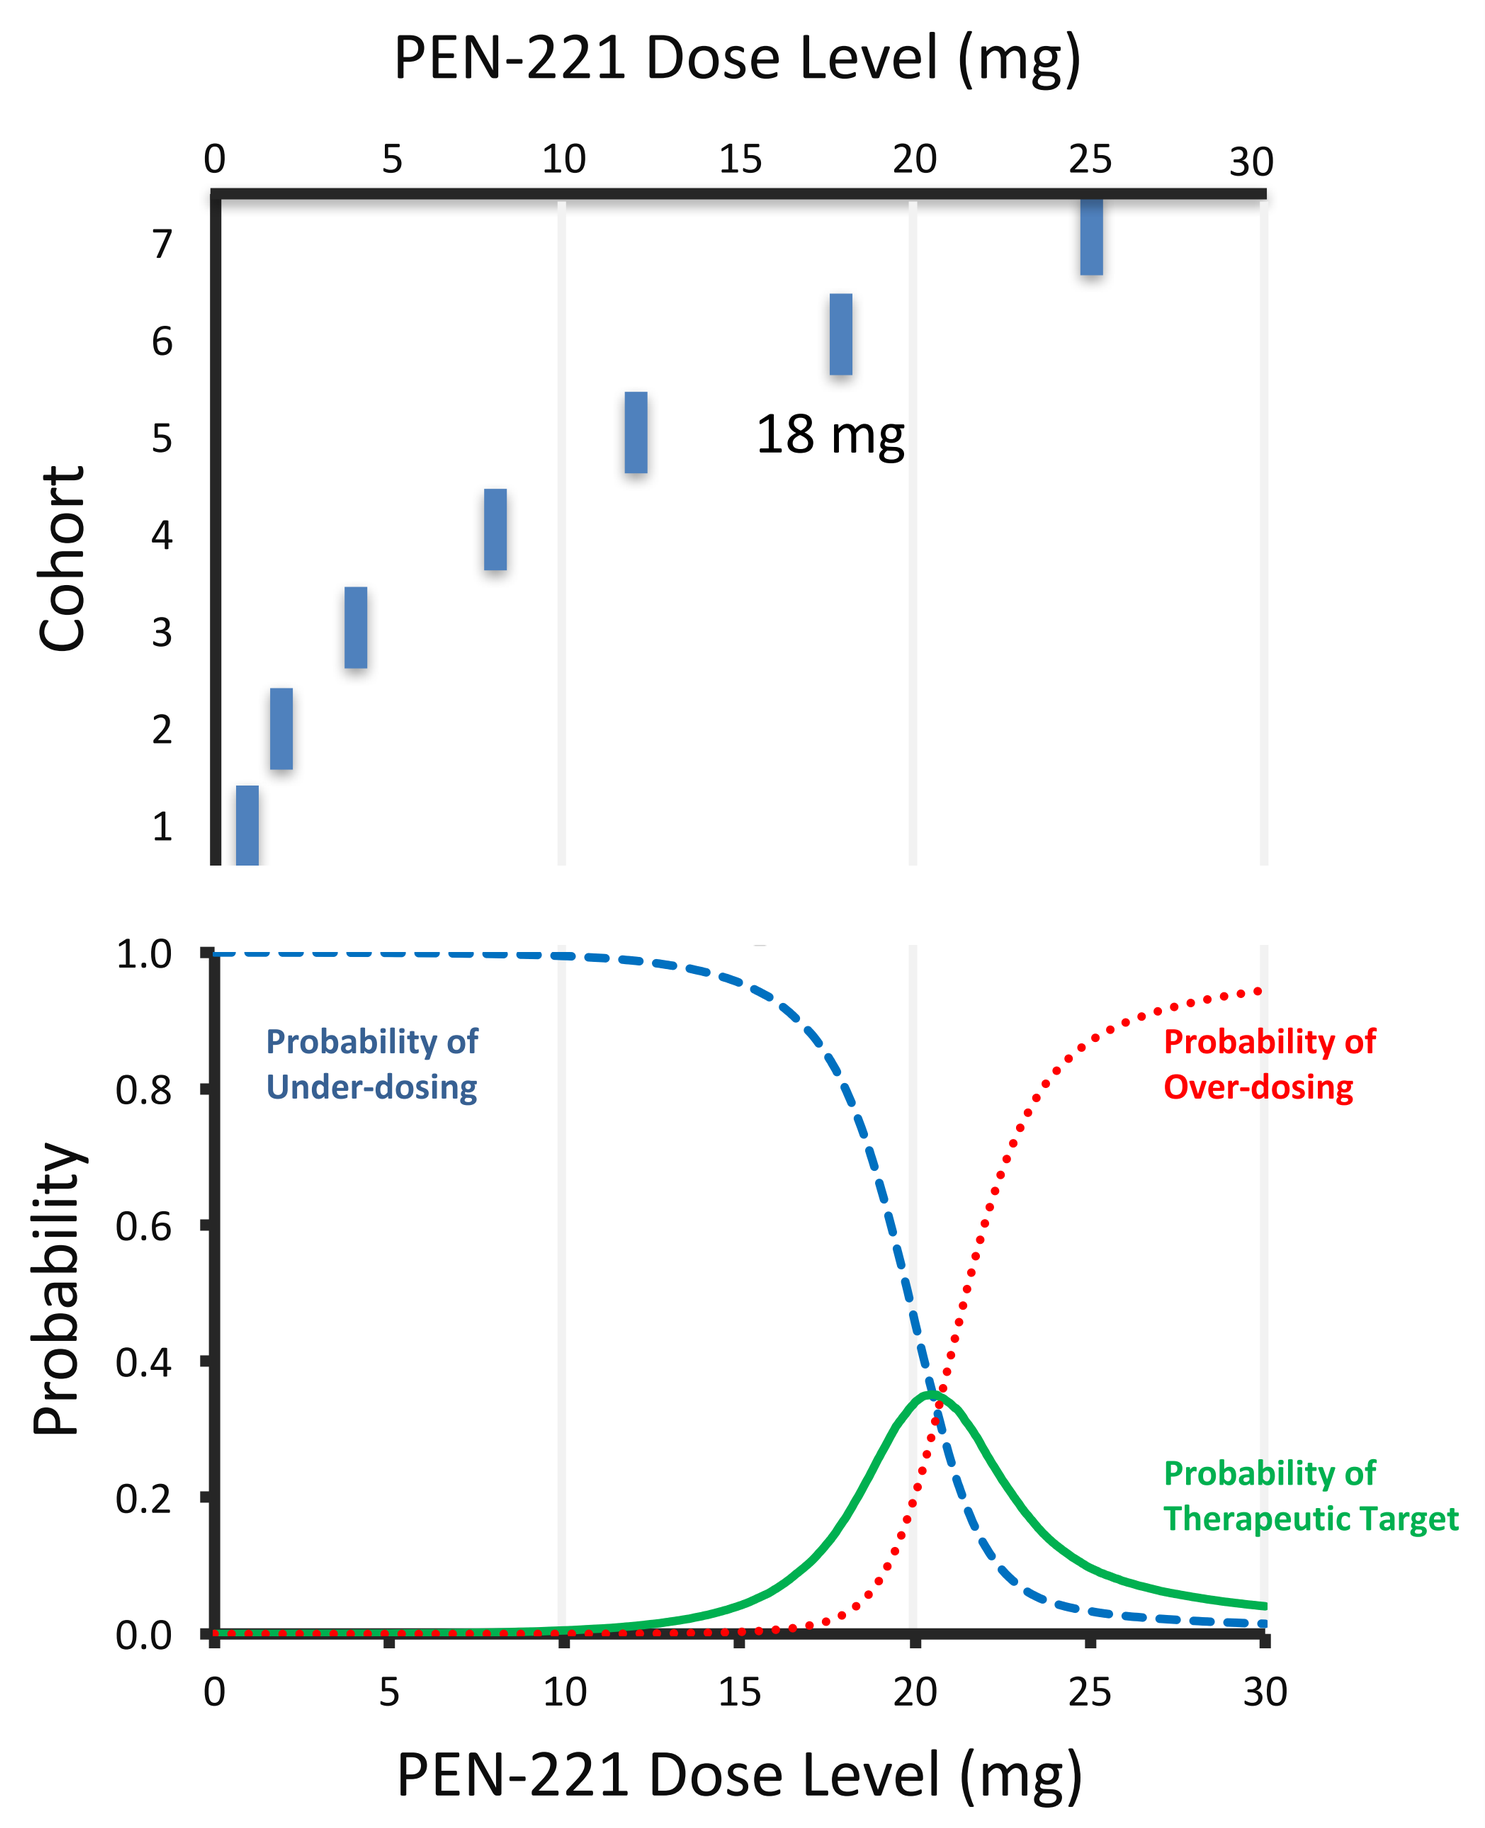
PEN-221 Dose Level (mg)
Cohort
18 mg
Probability
Probability of Under-dosing
Probability of Over-dosing
Probability of Therapeutic Target
PEN-221 Dose Level (mg)

- The dosing of PEN-221 was flat, starting with a Cohort 1 dose of 1mg
- There were 7 Cohorts with the highest being 25mg
- The MTD was declared to be 18mg after completing dosing of 6 patients at that level. The BLRM estimated a 35% chance that the probability of toxicity at 18mg lies between 16% and 33%, higher than any other dose fulfilling the EWOC criterion (Data up to 23-Feb-2018)

PEN-221 Treatment-Emergent Treatment-Related Adverse Events In ≥10% Of Patients On Study

Data up to 23-Feb-2018	Total Number (%)	Grade 1 Number (%)	Grade 2 Number (%)	Grade 3 Number (%) [Dose level]	Grade 4 Number (%)
Fatigue	11 (48)	5 (22)	6 (26)	0	0
Nausea	11 (48)	7 (30)	4 (17)	0	0
Diarrhoea	10 (44)	7 (30)	3 (13)	0	0
Neuropathy, peripheral	6 (26)	4 (17)	1 (4)	1 (4)[25mg]	0
Abdominal pain	6 (26)	3 (13)	3 (13)	0	0
Vomiting	6 (26)	5 (22)	1 (4)	0	0
ALT increase	5 (22)	3 (13)	1 (4)	1 (4)[25mg]	0
Decreased appetite	5 (22)	3 (13)	2 (9)	0	0
AST increase	4 (17)	2 (9)	1 (4)	1 (4)[25mg]	0
Constipation	4 (17)	2 (9)	1 (4)	1 (4)[2mg]	0
Dysgeusia	3 (13)	2 (9)	1 (4)	0	0
Myalgia	3 (13)	2 (9)	1 (4)	0	0
Pruritus	3 (13)	2 (9)	1 (4)	0	0

Additional PEN-221 Treatment-Emergent Treatment-Related Adverse Events With At Least 1 Grade 3 Or 4 Event

Data up to 23-Feb-2018	Total Number (%)	Grade 1 Number (%)	Grade 2 Number (%)	Grade 3 Number (%) [Dose level]	Grade 4 Number (%) [Dose level]
Mucosal inflammation	2 (9)	0	1 (4)	1 (4)[25mg]	0
Hyperglycemia	1 (4)	0	0	1 (4)[18mg]	0
Dermatitis	1 (4)	0	0	1 (4)[25mg]	0
Febrile Neutropenia	1 (4)	0	0	0	1 (4)[25mg]
Drug induced liver injury	1 (4)	0	0	1 (4)[25mg]	0
Infusion related reaction	1 (4)	0	0	1 (4)[4mg]	0

Plasma Pharmacokinetics

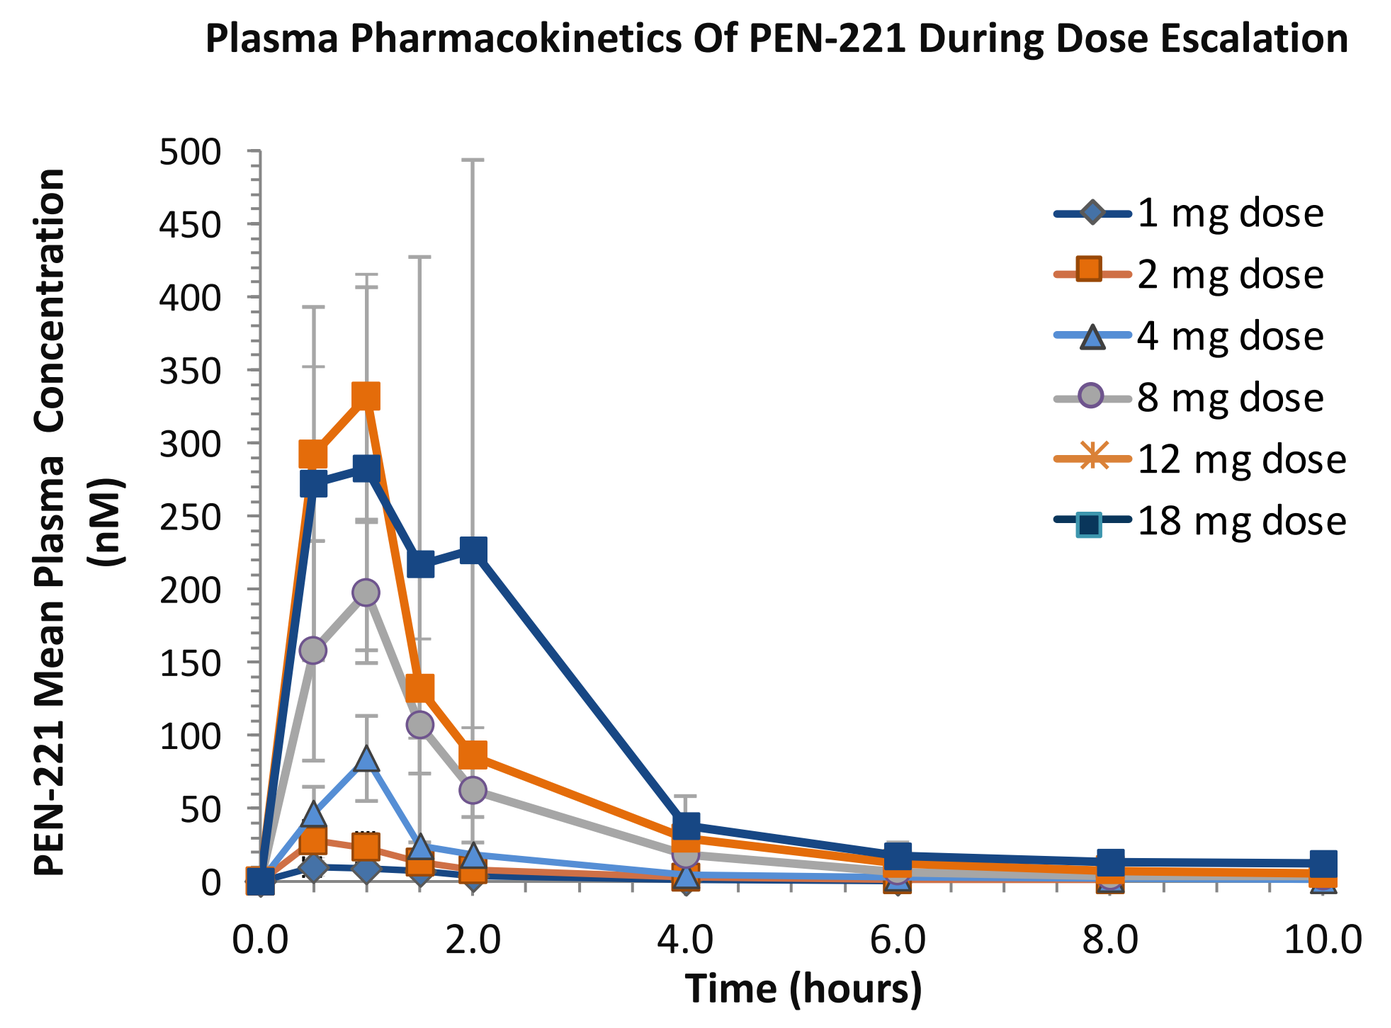
Plasma Pharmacokinetics Of PEN-221 During Dose Escalation
PEN-221 Mean Plasma Concentration (nM)
1 mg dose
2 mg dose
4 mg dose
8 mg dose
12 mg dose
18 mg dose
Time (hours)

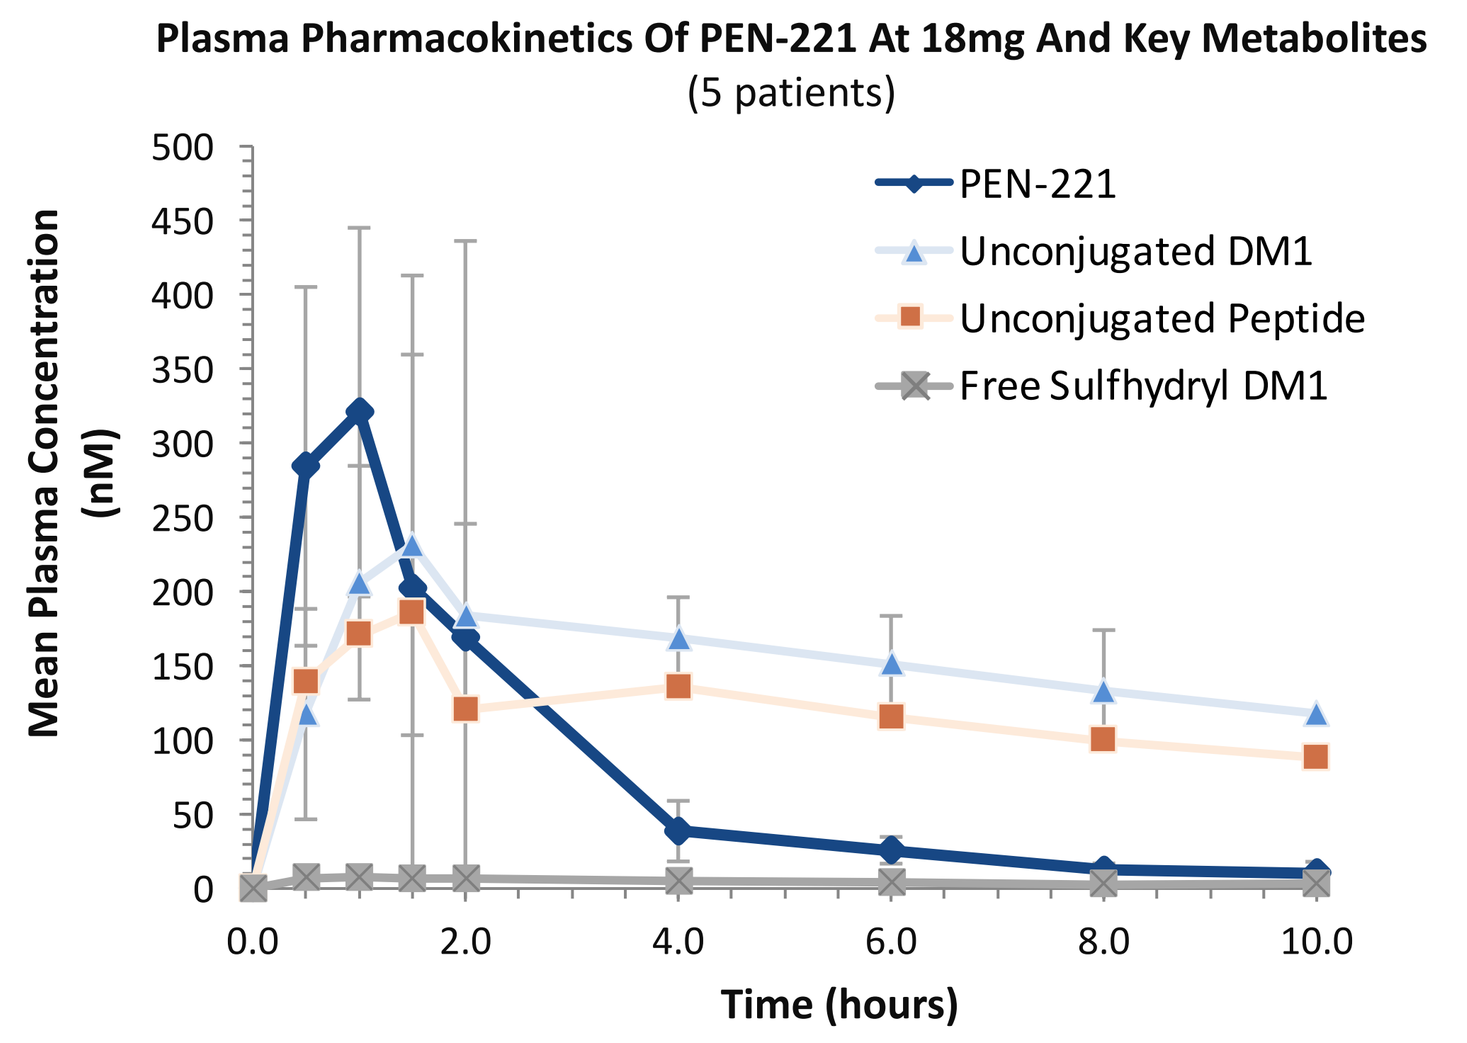
Plasma Pharmacokinetics Of PEN-221 At 18mg And Key Metabolites
(5 patients)
Mean Plasma Concentration (nM)
PEN-221
Unconjugated DM1
Unconjugated Peptide
Free Sulfhydryl DM1
Time (hours)

Mean PEN-221 Plasma Pharmacokinetics

PEN-221 Dose (mg)	Cmax (nM)	AUCinf (nM·h)	CL (L/h)	Vz (L)
1	7.47	14.6	38	65
2	20.5	37.9	35	80
4	74.3	118	20	60
8	186	341	15	37
12	293	584	13	33
18	381	666	16	36
25	741	1310	11	50

- PEN-221 concentration increased during the infusion with a median time of C_{max} ranged from 0.5 to 1.1 hours
- Median $t_{1/2}$ was 1.7 hours across all dose cohorts
- PEN-221 AUC_{inf} increased with the dose
- For the 12 mg and higher dose cohorts the AUC_{inf} was above efficacious exposure threshold seen in pre-clinical xenograft studies of SCLC models
- DM1-SH levels remained low with respect to PEN-221

Decrease In Circulating Biomarkers Of Neuroendocrine Disease, Chromogranin A (A), Neuron Specific Enolase(B) And Circulating Tumor Cells (C), In A Patient With A GI NET Treated With PEN-221

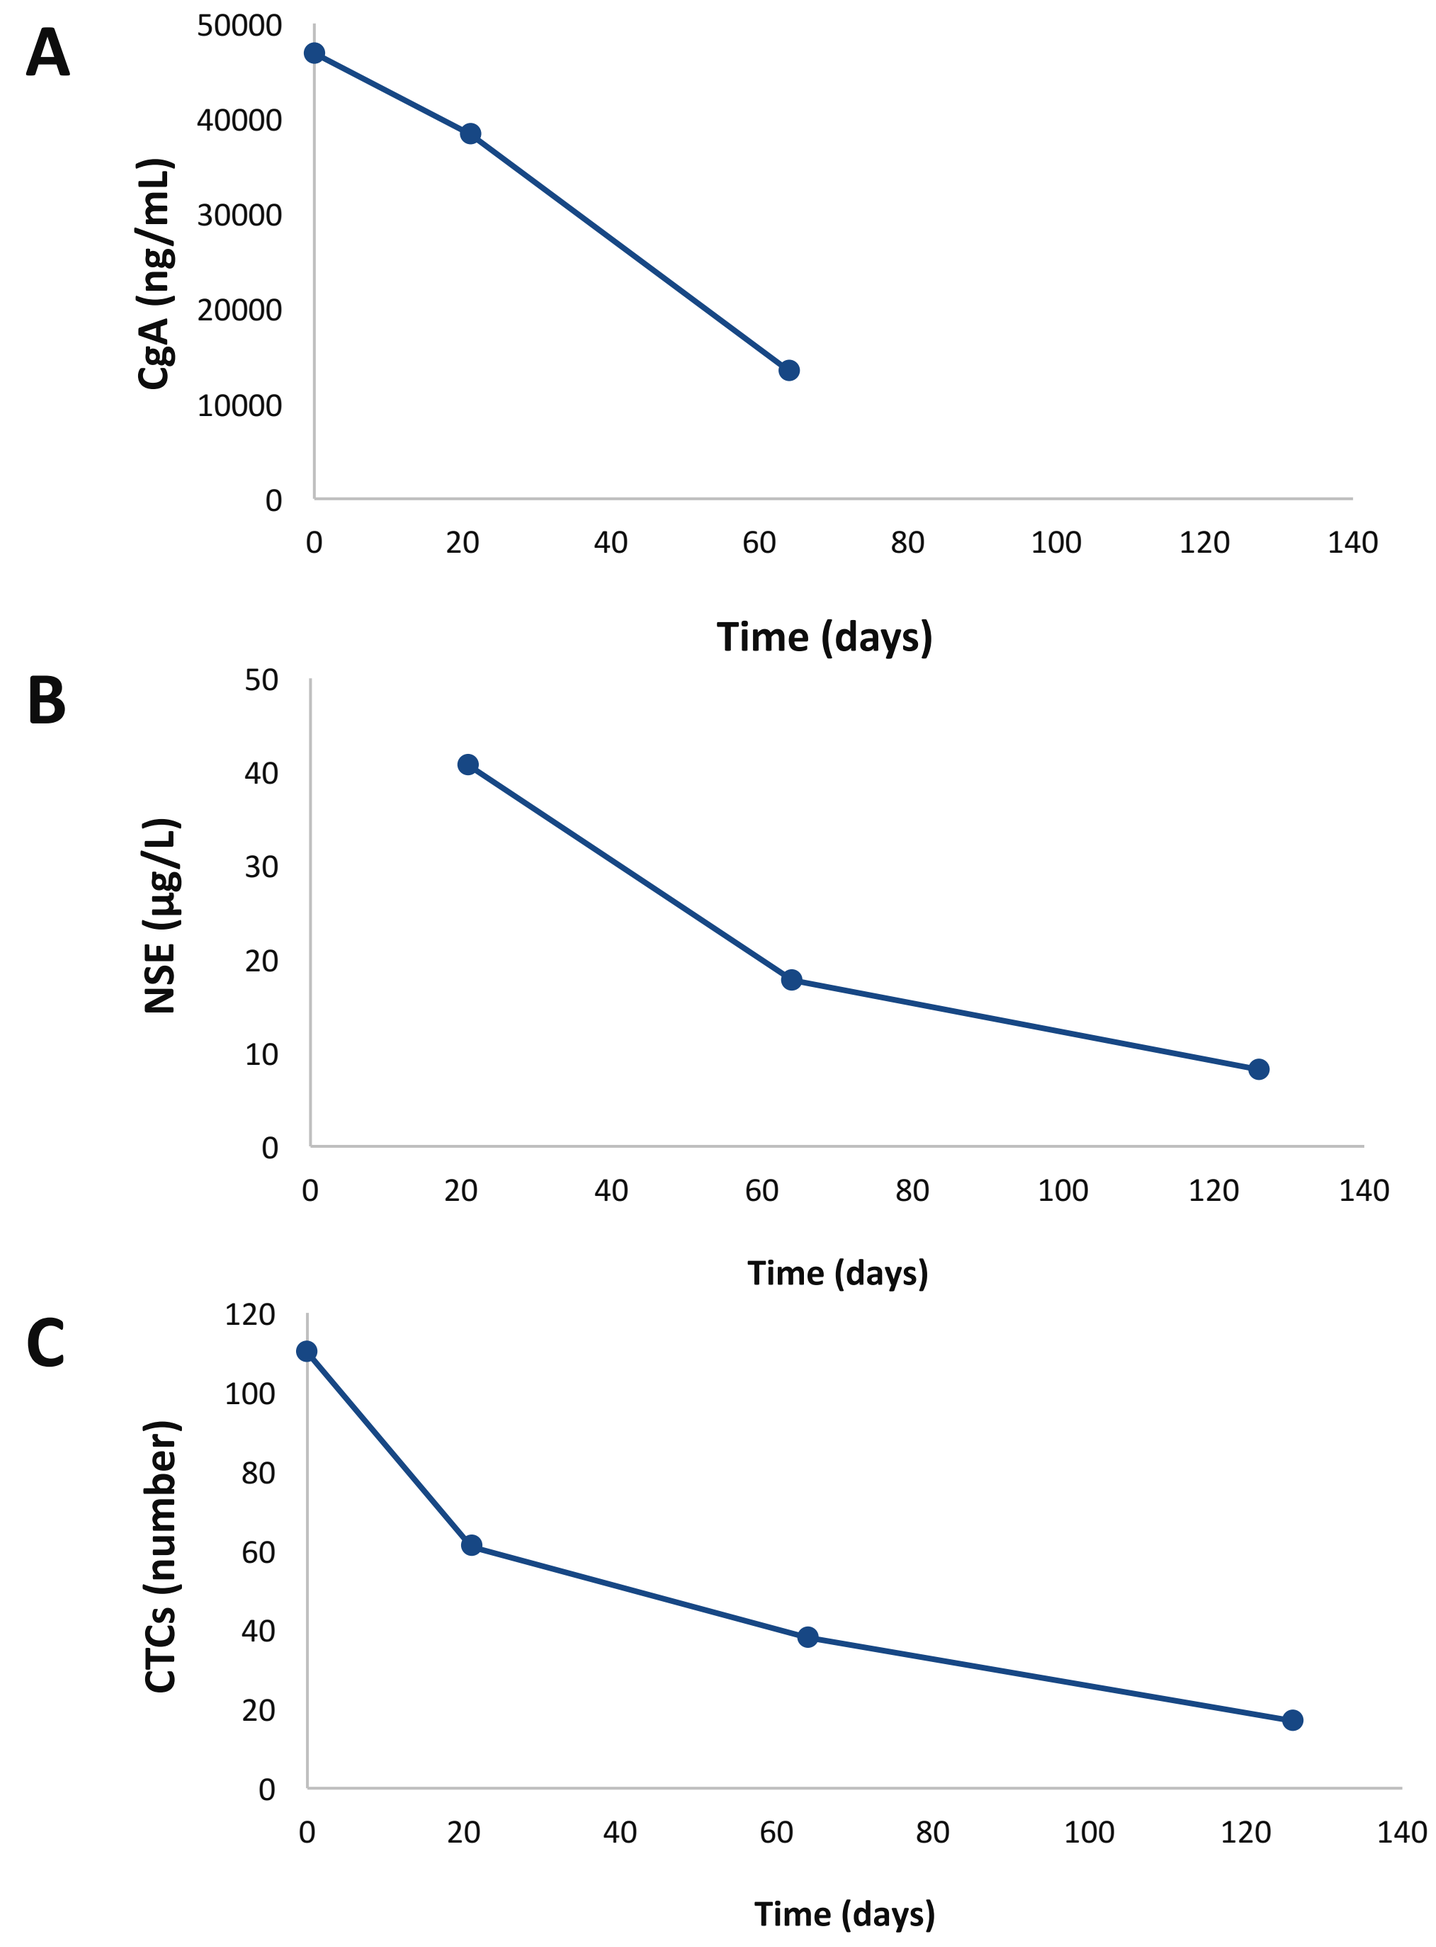
A
CgA (ng/mL)
Time (days)
B
NSE (µg/L)
Time (days)
C
CTCs (number)
Time (days)

- Circulating biomarker relevant to neuroendocrine tumors were selected:
 - CgA was predictive of morphological response, PFS, and OS in pancreatic NETs (Yao et al)
 - Decline in plasma concentration of NSE has been associated with response to treatment (Isgro et al)
 - Changes in CTCs has been associated with response to treatment in patients with NETs (Khan et al)

- 45 year old Female with well differentiated, intermediate grade GI (large bowel) NET with metastasis to lymph nodes, bone, lung, and brain.
- Progressed on all prior therapies: lanreotide, mTOR inhibitor, 177Lu-DOTATATE
- Receiving PEN-221, 18 mg
- Disease related symptoms of flushing disappeared after first dose and have not recurred
- Chromogranin A (CgA, A), Neuron Specific Enolase (NSE, B) and Circulating Tumor Cells (CTCs, C) were measured over time
- These 3 biomarkers markedly decreased over time

Duration Of Treatment As Of 23-Feb-2018

- Minor tumor responses were seen in 3 of the 7 NET patients (GI or pancreatic) who received 8 mg – 18 mg of PEN-221
- Duration of Stable Disease is a key measure of benefit for patients with NETs

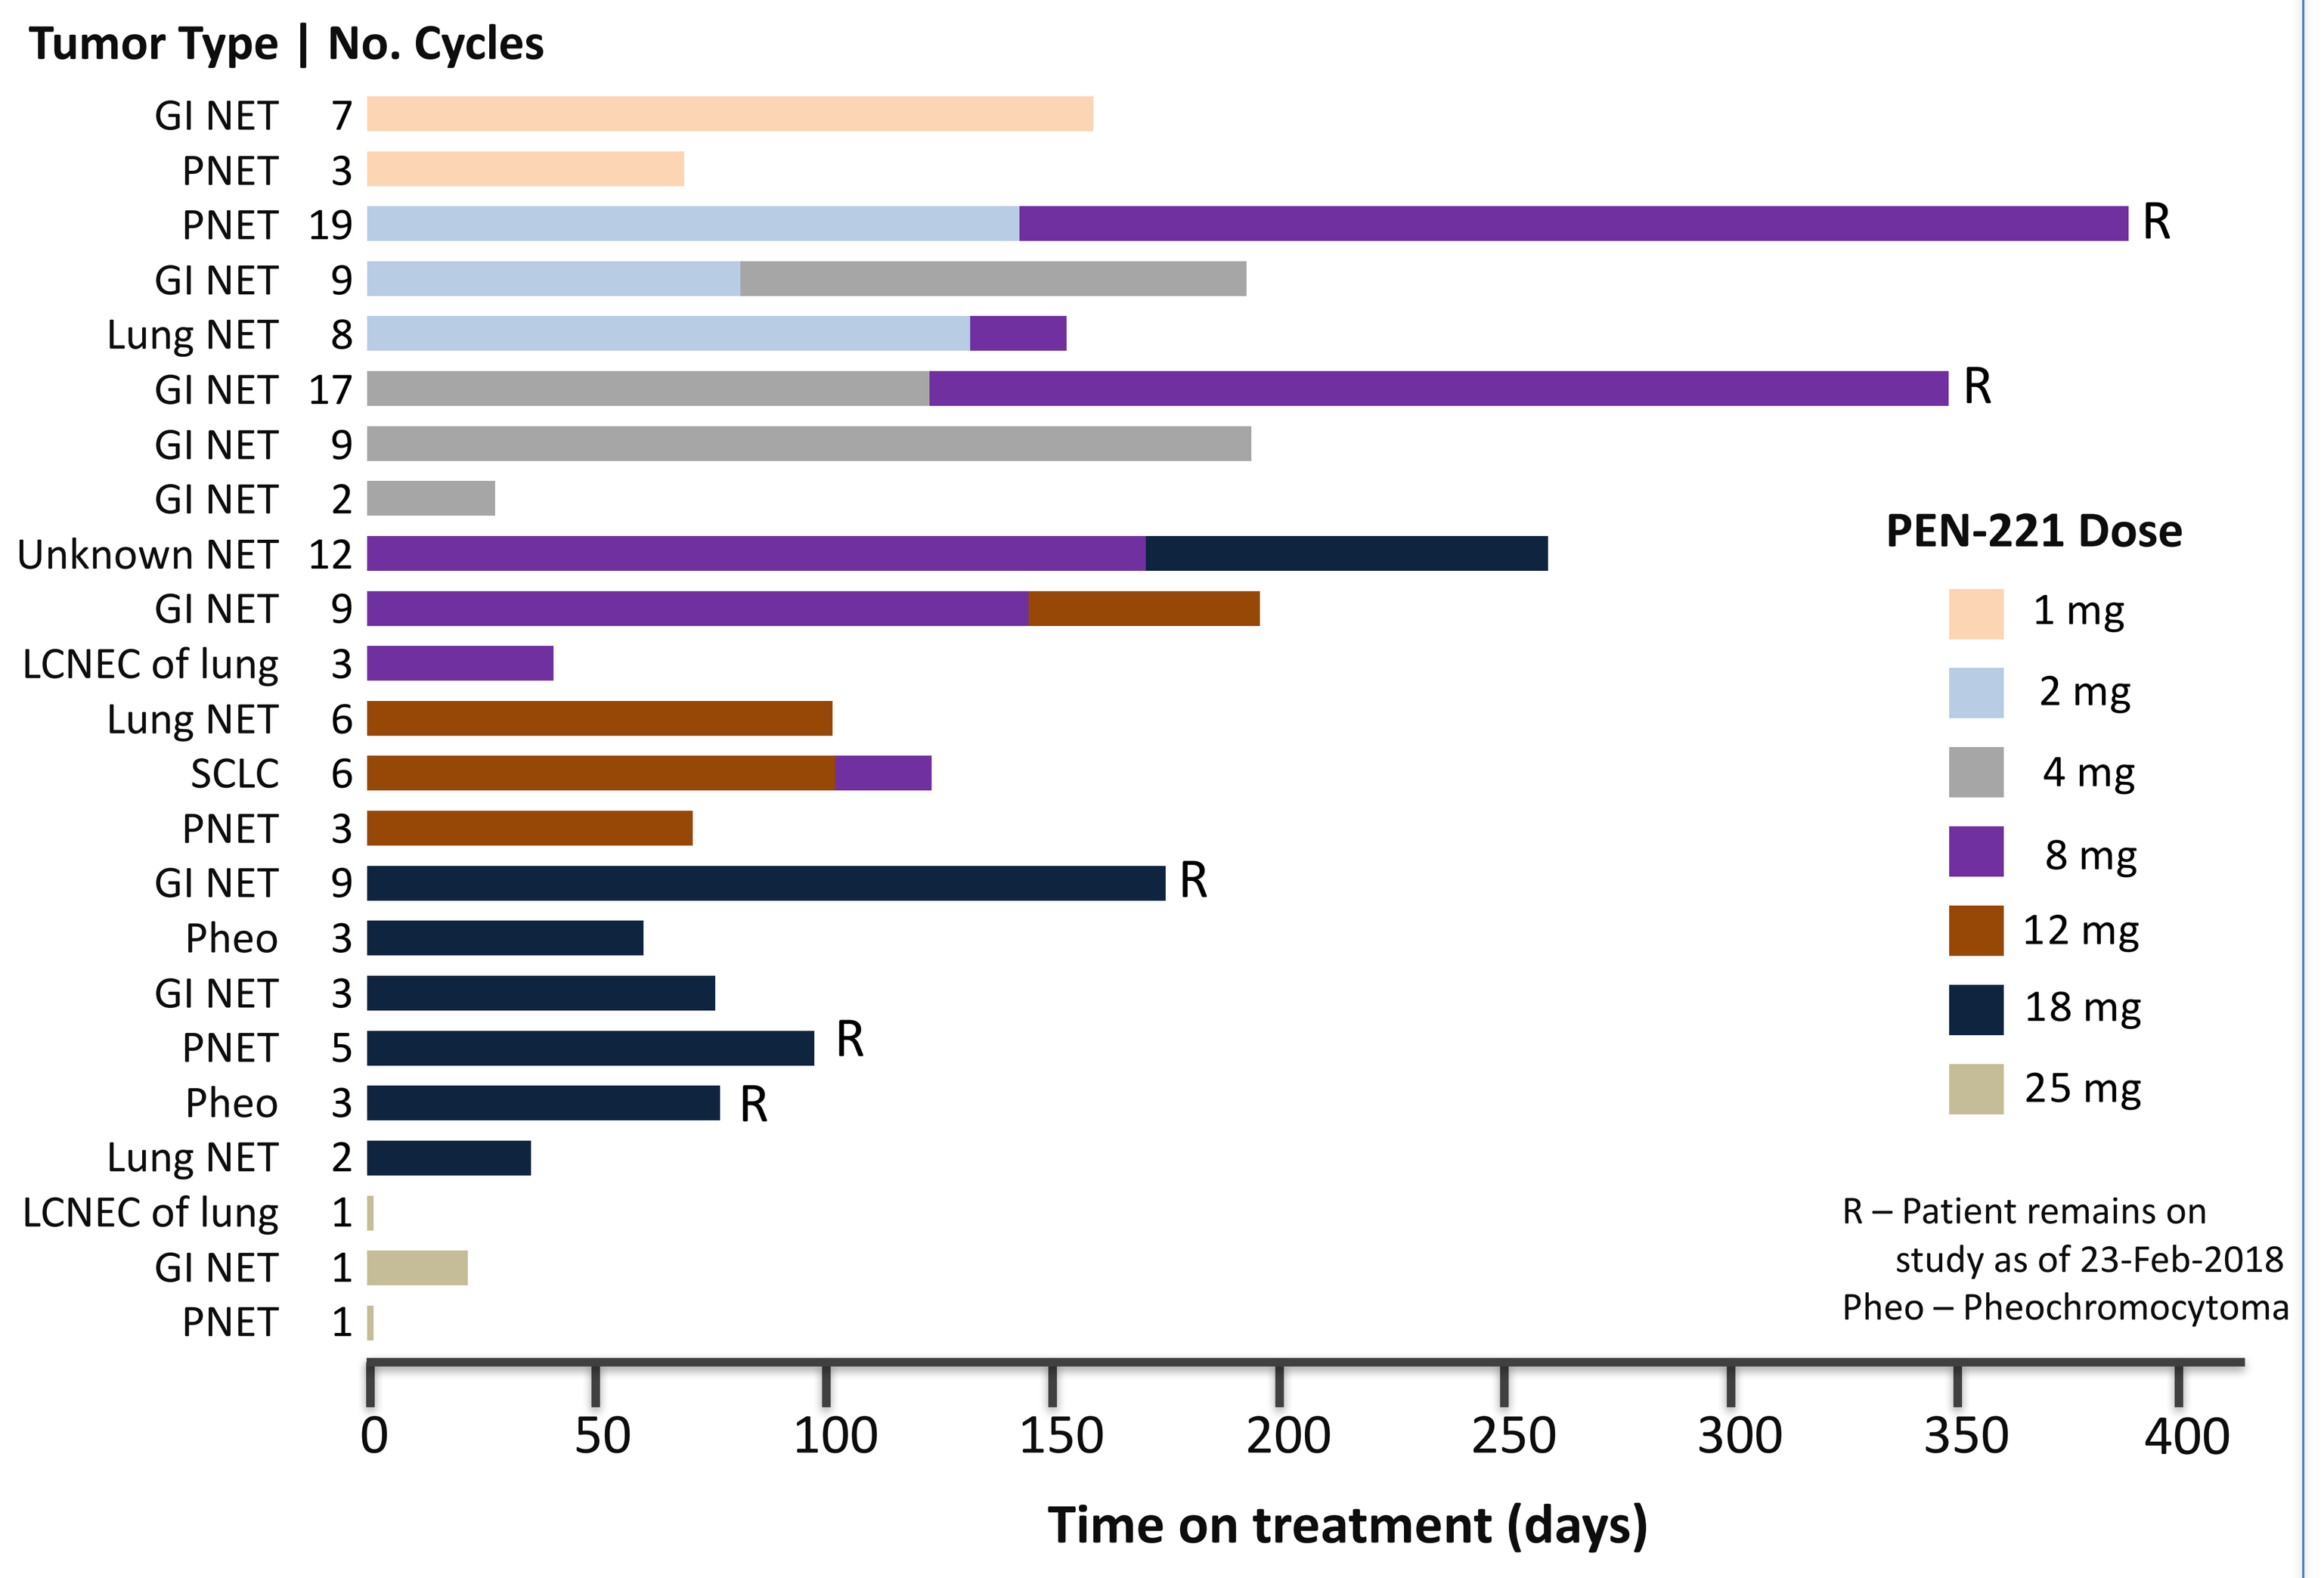
Tumor Type | No. Cycles
GI NET 7
PNET 3
PNET 19 R
GI NET 9
Lung NET 8
GI NET 17 R
GI NET 9
GI NET 2
Unknown NET 12
GI NET 9
LCNEC of lung 3
Lung NET 6
SCLC 6
PNET 3
GI NET 9 R
Pheo 3
GI NET 3
PNET 5 R
Pheo 3 R
Lung NET 2
LCNEC of lung 1
GI NET 1
PNET 1
PEN-221 Dose
1 mg
2 mg
4 mg
8 mg
12 mg
18 mg
25 mg
R – Patient remains on study as of 23-Feb-2018
Pheo – Pheochromocytoma
Time on treatment (days)

Conclusions

- PEN-221 appears well tolerated with no DLTs in the first 6 cohorts (1-18 mg; 20 pts). In cohort 7 (25mg), 2 of 3 pts had DLTs that rapidly and fully resolved
- The MTD and RP2D of PEN-221 was established at 18 mg
- The plasma exposure of PEN-221 increased with dose, median $t_{1/2}$ ~1.7 h.
- Doses of ≥12mg PEN-221 give an exposure above the efficacious threshold seen in pre-clinical xenograft SCLC models
- As of 23-Feb-2018 there was preliminary evidence of antitumor activity:
 - One patient had a rapid and sustained decrease in chromogranin A, Neuron Specific Enolase and circulating tumor cells
 - Among 15 NET pts who were evaluable for response, 11 had stable disease (SD) at 9 weeks, of whom 8 were sustained for 18 – 45 weeks, including 2 ongoing patients with SD for 44 and 45 weeks.
 - Target lesion shrinkage leading to minor responses were observed in 3/7 patients who had either a GI or pancreatic NET (dose range 8-18 mg)
 - The one SCLC patient on the study had SD for 12 weeks
- PEN-221 (18 mg q 3 weeks) is being evaluated in Phase 2a expansion cohorts enrolling midgut NET, pancreatic NET, and SCLC patients (ClinicalTrials.gov Identifier: NCT02936323)

Update

- As of 11-Apr-2018, 4 patients remain on study with SD of 5, 7, 12, 14 months respectively

We thank the patients who enrolled in this study and their families

References

Babb et al 1998 Stat Med 1998;17:1103-20.

Neuenschwander et al 2008 Stat Med 2008;27:2420-39

Yao JC et al, 2011, Journal of Clinical Endocrinology and Metabolism 96:3741–3749

Isgro et al, 2015, Adv Exp Med Biol. 867:125-43

Khan et al, 2016, Clin Cancer Res. 22:79-8